Evaluating the Value of Clinical and Economic Evidence-Based Information from Dossier Submissions Using the Academy of Managed Care Pharmacy (AMCP) Format

Rowell D. Medina, PharmD[1,2]; Divyesh Khetia, PharmD[1]; Purnima J. Topiwala, PharmD[3]; Vincent J. Willey, PharmD[2]; Gary E. Sloskey, PharmD, BCPS[2]; Wayne Langholff, PhD[4]

[1]Centocor Ortho Biotech Services, LLC; [2]Philadelphia College of Pharmacy, University of the Sciences; [3]Ortho-McNeil Janssen Scientific Affairs, LLC; [4]Johnson and Johnson Pharmaceutical Research & Development

BACKGROUND

- AMCP Format for Formulary Submissions was designed to achieve two goals.[1,2]
 1. Improve timeliness, scope, quality, and relevance of information
 2. Streamline the process of acquiring data and reviewing products
- Pharmaceutical manufacturers use this format as a tool to standardize and communicate evidence on the clinical impact and economic value of products when responding to unsolicited requests from payers.[1,2]
- The latest version of the AMCP Format was released in October 2009.
- Previous studies focused on formulary inclusion, dossier quality, organizational burden, and impact of pharmacoeconomic models with limited research comparing the perspectives of payers and manufacturers specific to clinical and economic value.[3,4,5,6,7,8]
- Although dossier submissions continue to be widely accepted, it is unknown whether clinical evidence or economic evidence is more useful to pharmacy and therapeutics (P&T) committees when making informed formulary decisions.

OBJECTIVE

- To evaluate and compare the perspectives of payers (managed care organizations or pharmacy benefit managers) and pharmaceutical manufacturers regarding specific sections of the AMCP Format for Formulary Submissions, Version 3.0:
 - Section 3: Supporting Clinical Evidence
 - Section 4: Economic Value and Modeling Reports

METHODS

- Study design: multi-centered, prospective, cross-sectional survey
- Two anonymous surveys were developed using Inquisite™. This is a web-based survey program that allows the surveyor to manage data collection.
 - Both surveys were intentionally structured so that many of the questions would mirror each other to make side-by-side comparisons.
 - A four-point Likert scale was used to avoid a "neutral" response choice.
- Recipients were asked to complete their respective survey only if they were familiar with the review process or preparation methods of AMCP dossier submissions in 2010.
- The surveys were available for two weeks from January 17, 2011 through January 31, 2011.

Payer Survey

- A 22-question electronic survey was disseminated via e-mail to 65 payers representing their P&T committee or its equivalent.
- Payers were selected from:
 - Pharmacy Benefit Management Institute Directory
 - AMCP list of managed care pharmacy residency programs
 - Recruitment during networking events (i.e. residency showcases, career fairs)
- The survey consisted of a series of questions based on their professional opinion of the value, helpfulness, and completeness of the evidence-based information in Sections 3 and 4 of the AMCP Format.
- Survey questions included the following:
 - 2Q: Demographics
 - 7Q: Section 3 (Supporting Clinical Evidence)
 - 11Q: Section 4 (Economic Value and Modeling Reports)
 - 2Q: Overall opinions (open-ended)

Pharmaceutical Manufacturer Survey

- A 21-question electronic survey was distributed via e-mail to a Medical Information (MI) representative from 40 pharmaceutical manufacturers.
- The questions were geared to the preparation methods, time investments, and concerns of creating a dossier.
- Survey questions included the following:
 - 1Q: Demographics
 - 5Q: Section 3 (Supporting Clinical Evidence)
 - 8Q: Section 4 (Economic Value and Modeling Reports)
 - 2Q: Confidentiality concerns
 - 5Q: Dossier submission request trend (last 5 years)

Abbreviation: Q = questions

RESULTS

Demographics: Payers (n=12)

- Of 65 managed care organization and pharmacy benefit managers contacted, 23 (35%) responded.
 - Of the 65 payers, 12 (18%) completed the survey.
 - 17% (11/65) responded but did not complete the survey.
 - Declined participation (n=1)
 - Not permitted to participate in external surveys (n=8)
 - Did not accept dossiers (n=2)
- The majority of the respondents (58%, 7/12) were pharmacy directors [Figure 1].
- Half identified their company as a large payer, handling greater than one million lives [Figure 2].

Figure 1. Respondent Background

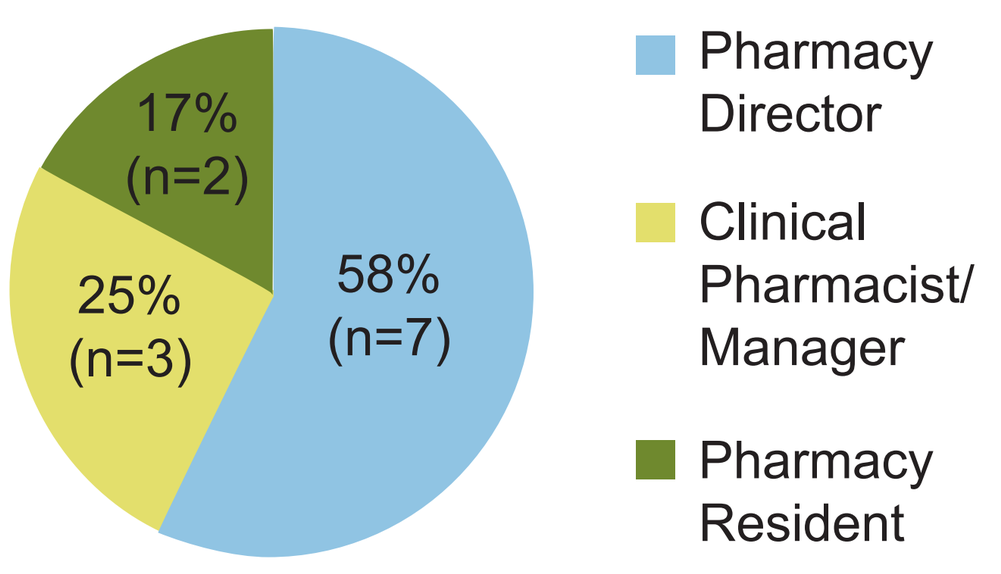
17% (n=2)
25% (n=3)
58% (n=7)
Pharmacy Director
Clinical Pharmacist/ Manager
Pharmacy Resident

Figure 2. Payer Size

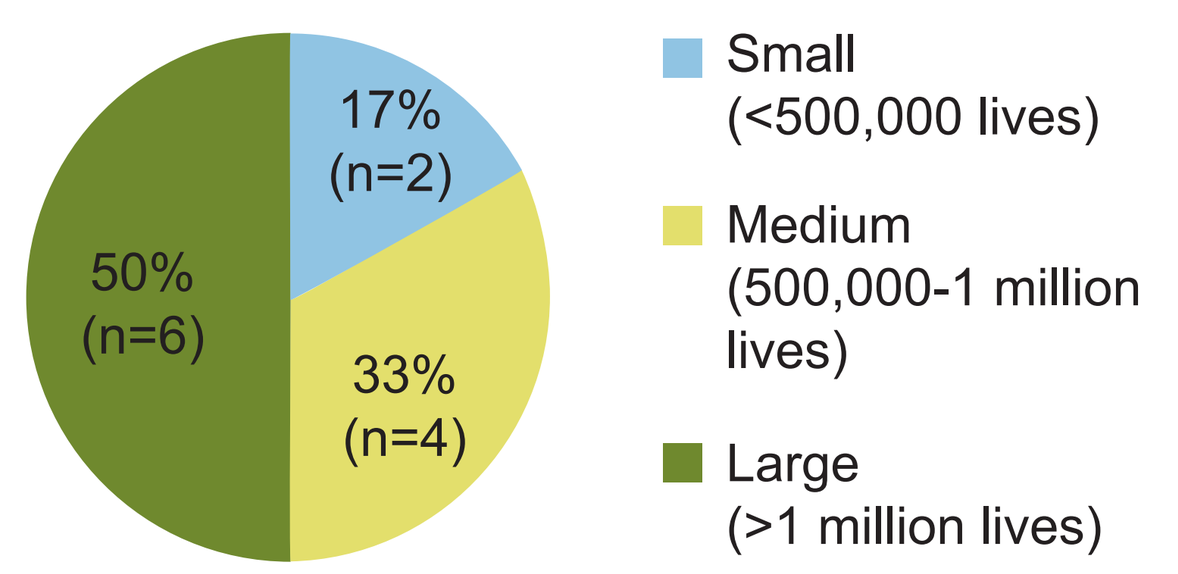
17% (n=2)
50% (n=6)
33% (n=4)
Small (<500,000 lives)
Medium (500,000-1 million lives)
Large (>1 million lives)

Demographics: Pharmaceutical Manufacturers (n=13)

- The mean number of drug product dossiers that was supported by each Medical Information department was 8 (range 2-25) [Figure 3].
- Generally, the number of dossier requests has been increasing linearly [Figure 4].

Figure 3. Number of Drug Product Dossiers Supported by Manufacturers

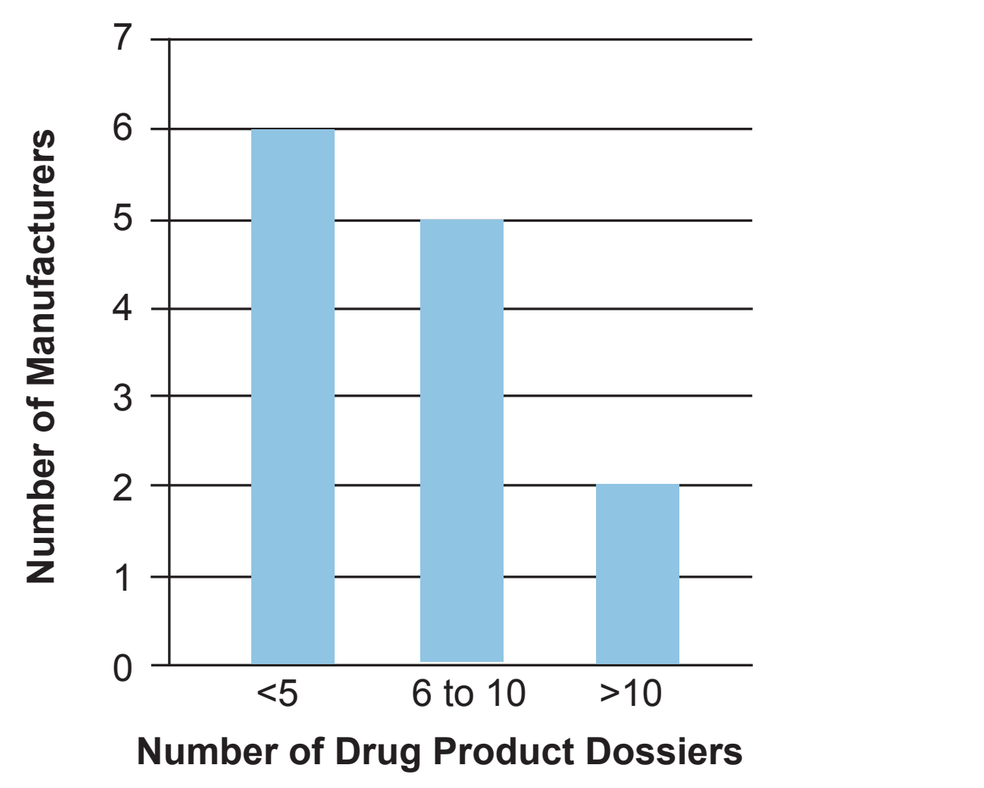
Number of Manufacturers
<5
6 to 10
>10
Number of Drug Product Dossiers

Figure 4. Trends of Dossier Requests (n=11)

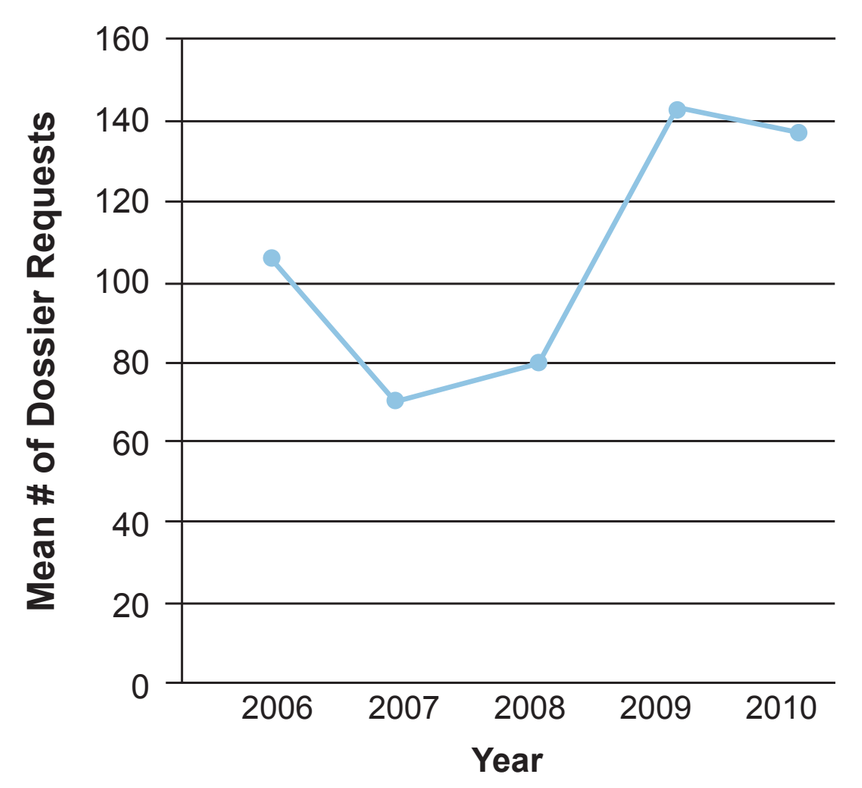
Mean # of Dossier Requests
2006
2007
2008
2009
2010
Year

Section 3: Supporting Clinical Evidence

- Published trials for labeled indications [Figure 5] were considered "valuable" to payers with a mean of 2.25 and were "always included" in dossiers by all 13 manufacturers.
- Half of the payers (n=6) considered unpublished trials for labeled indications as only "somewhat valuable" [Figure 6], while 46% of manufacturers "always included" those studies in dossiers.

Table 1. Likert Scale Key*

3	Very valuable/Always included
2	Valuable/Often included
1	Somewhat valuable/Sometimes included
0	Not valuable/Not usually included

*Refer to Table 1 for Figures 5, 6, 7, and 8.

Figure 5. Labeled Indications: Published

Payers: 2.25 (mean)
Manufacturers: 3.00 (mean)

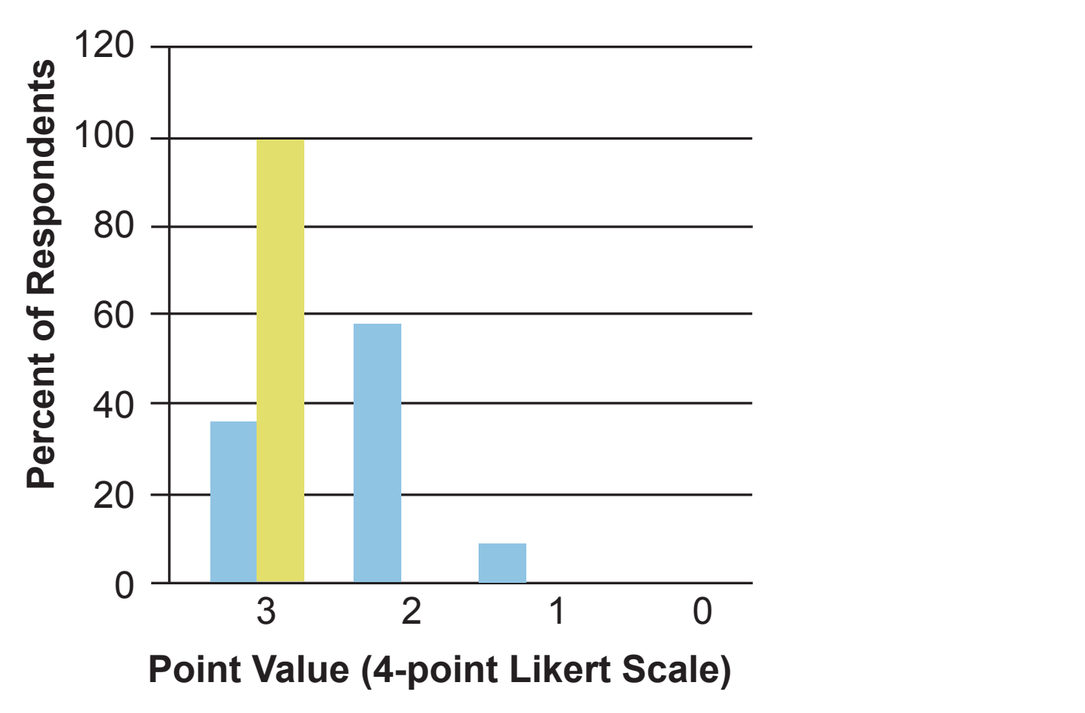
Percent of Respondents
Point Value (4-point Likert Scale)

Figure 6. Labeled Indications: Unpublished

Payers: 1.58 (mean)
Manufacturers: 2.15 (mean)

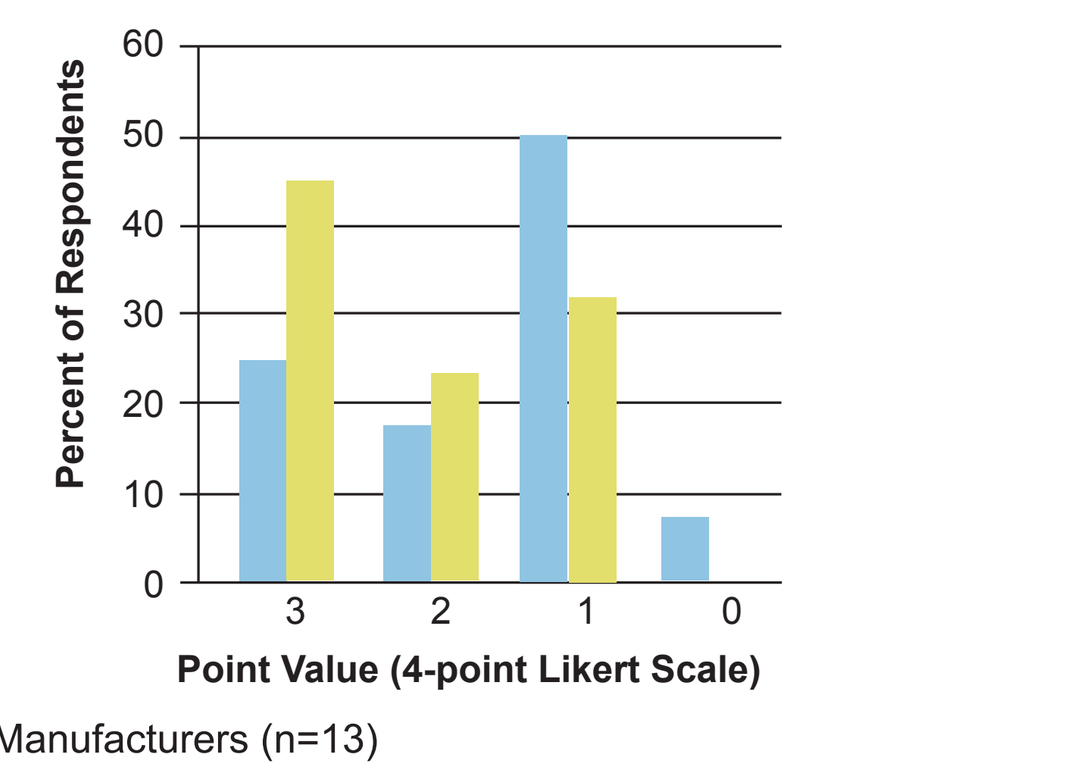
Percent of Respondents
Point Value (4-point Likert Scale)
Payers (n=12)
Manufacturers (n=13)

- Both payers and manufacturers had similar opinions on published trials for off-labeled indications [Figure 7] with a mean of 2.00 (valuable) and 2.08 (often included), respectively.
- 67% (8/12) of payers thought unpublished trials for off-labeled indications [Figure 8] were either "very valuable" or "valuable," but those studies were only "sometimes included" or "not usually included" by 77% (10/13) of manufacturers.

Figure 7. Off-Labeled Indications: Published

Payers: 2.00 (mean)
Manufacturers: 2.08 (mean)

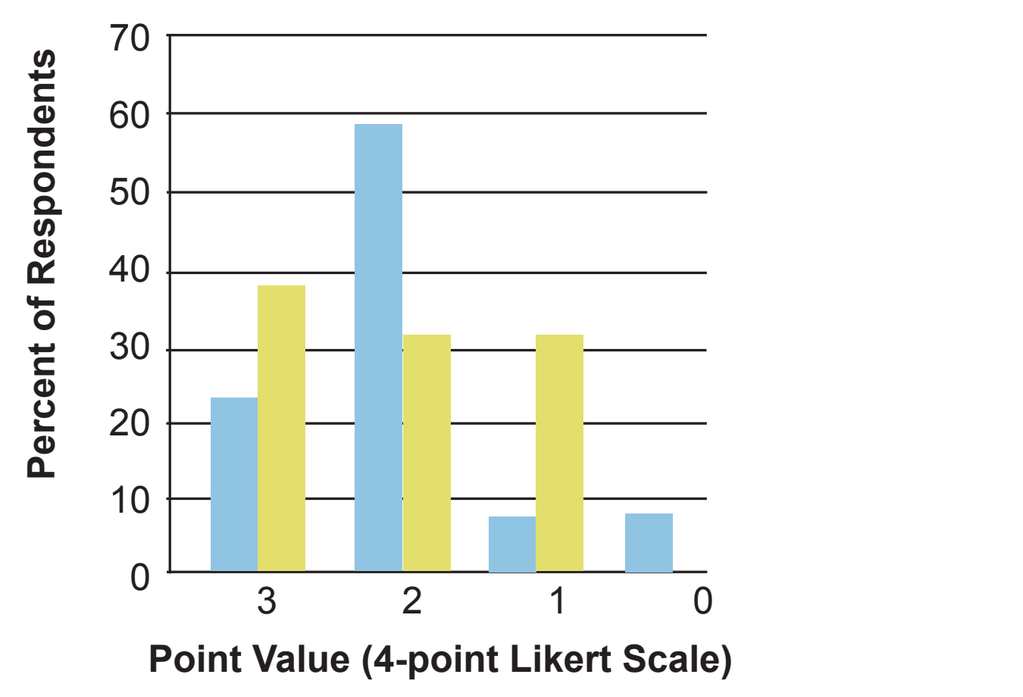
Percent of Respondents
Point Value (4-point Likert Scale)

Figure 8. Off-Labeled Indications: Unpublished

Payers: 1.67 (mean)
Manufacturers: 1.08 (mean)

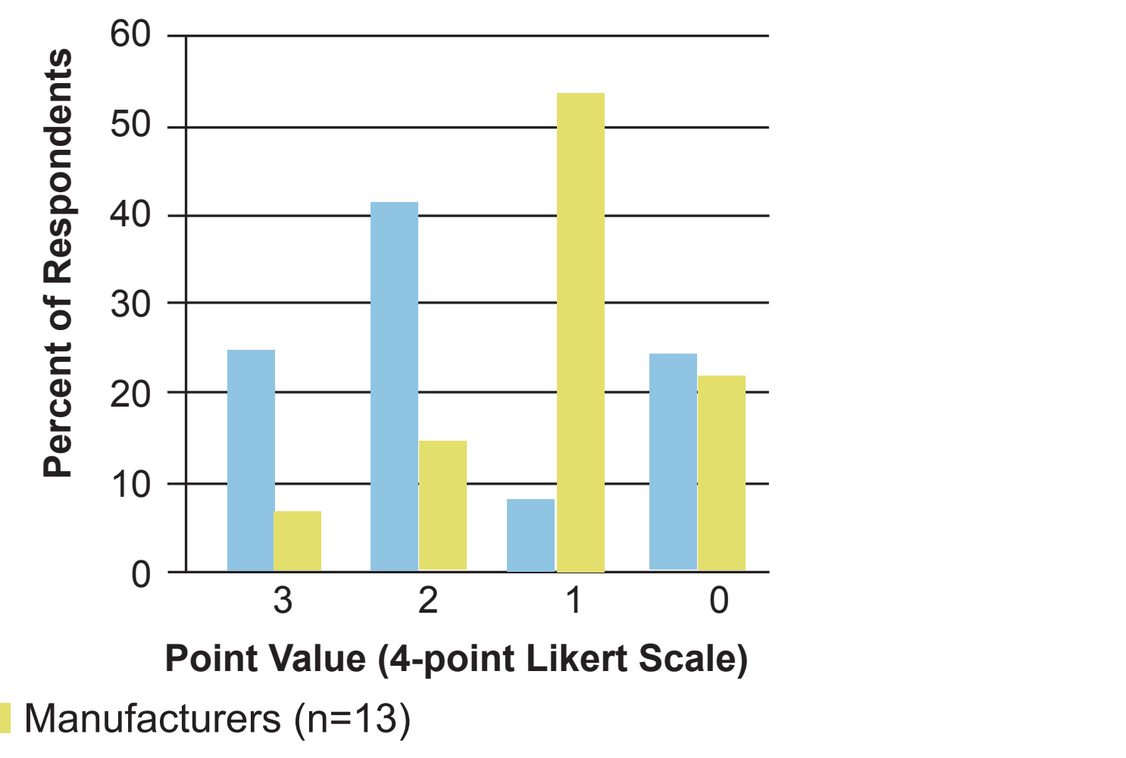
Percent of Respondents
Point Value (4-point Likert Scale)
Payers (n=12)
Manufacturers (n=13)

Section 4: Economic Value and Modeling Reports

- 17% (2/12) of payers did not review economic models, and 31% (4/13) of manufacturers did not include economic models in their dossier submissions.
- The majority of payers (70%, 7/10) and manufacturers (67%, 6/9) ranked cost effectiveness models first [Figure 9] in terms of value and information consistently included in the dossier.
- The budget impact models [Figure 10] were least valued by 60% (6/10) of payers, whereas financial models [Figure 11] were least included (ranked third) by 67% (6/9) of manufacturers.

Table 2. Ranking Key†

Ranked 1st	Most Valuable/Consistently included
Ranked 2nd	Valuable/Included
Ranked 3rd	Least Valuable/Not consistently included

†Refer to Table 2 for Figures 9, 10, and 11.

Figure 9. Cost Effectiveness Model

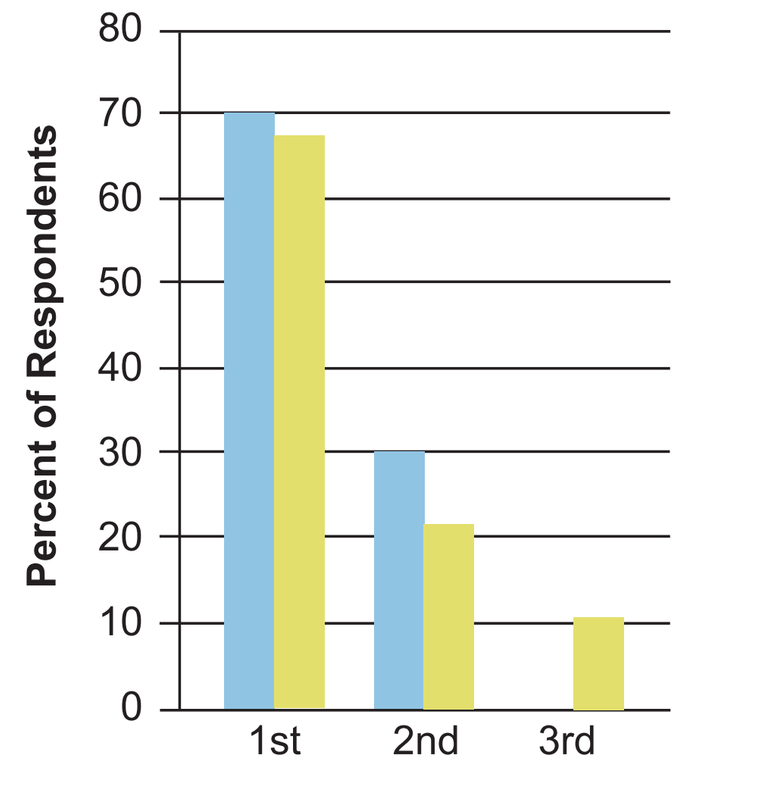

Figure 10. Budget Impact Model

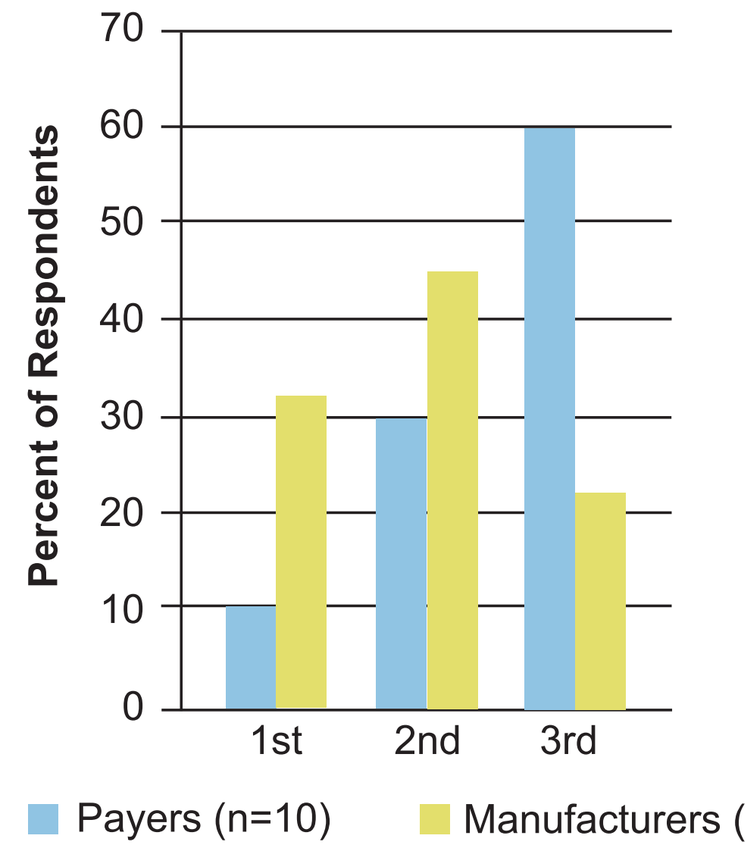

Figure 11. Financial Model

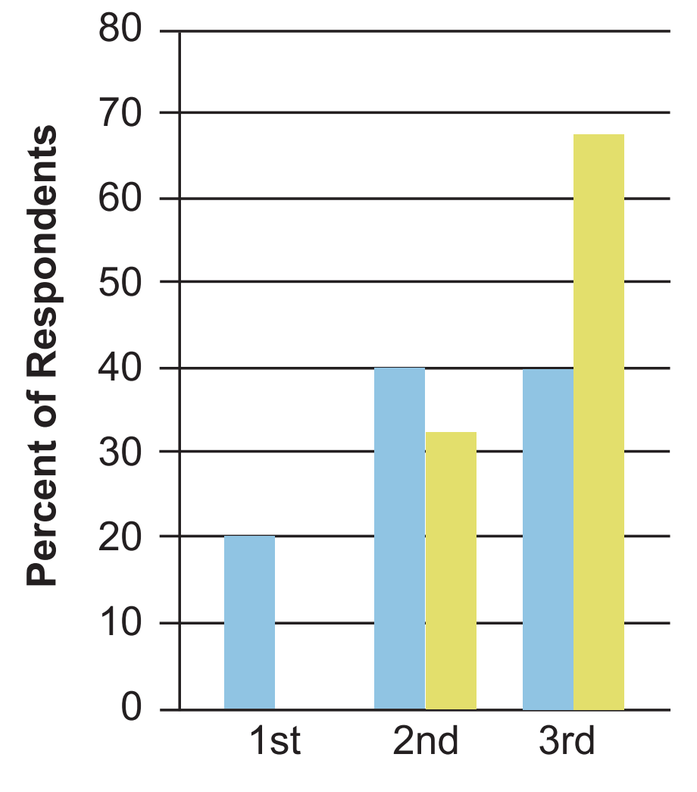
Percent of Respondents
1st
2nd
3rd
Payers (n=10)
Manufacturers (n=9)

- Of those who answered the modeling technique question when using cost effectiveness models, all eight payers valued decision trees the most, as opposed to only 33% (2/6) of manufacturers [Figure 12].
- Markov (cohort) models were considered the most valued technique among manufacturers (67%, 4/6).

Figure12. Modeling Technique Used when Constructing Cost Effectiveness Models

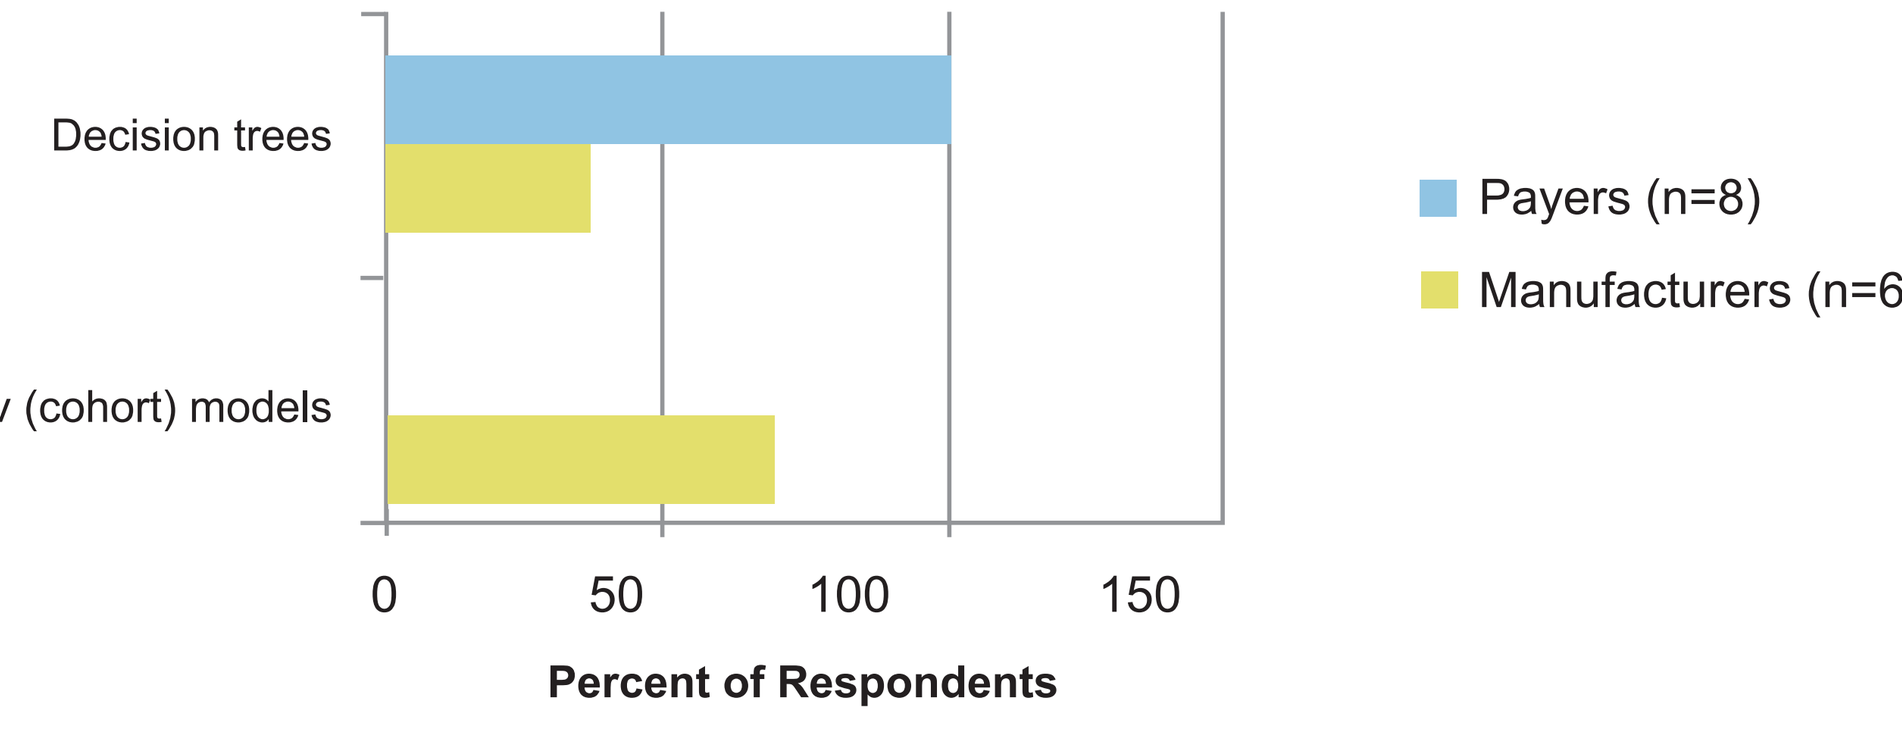
Decision trees
Markov (cohort) models
Payers (n=8)
Manufacturers (n=6)
Percent of Respondents

Ease of Use and Clarity

- Of those payers who reviewed economic models (n=10), they generally thought economic models were only "somewhat easy" to use (mean 1.20) and "somewhat clear" (mean 1.40) on a four-point Likert scale, 0-3.

Transparency

- The majority of payers (70%, 7/10) thought that the models were only "somewhat transparent" (mean 1.30) on a four-point Likert scale, 0-3.
- However, the nine manufacturers who included economic models in their drug product dossiers believed their models were transparent to "most" (89%, 8/9) or "all payers" (11%, 1/9) for a mean of 2.11.
- 67% (6/9) of manufacturers implemented their models in spreadsheet software.
 - The implementation of models in spreadsheet software is used to enhance transparency and ease of use.

Overall Opinions of Sections 3 & 4

Table 3. Point Scale Key‡

3 Point Scale	Very influential/Very complete/Very valuable
2 Point Scale	Influential/Complete/Valuable
1 Point Scale	Somewhat influential/Somewhat complete/Somewhat valuable
0 Point Scale	Not influential/Not complete/Not valuable

‡Refer to Table 3 for Figures 13 and 14.

Figure 13. Overall Payers' Opinions of Section 3

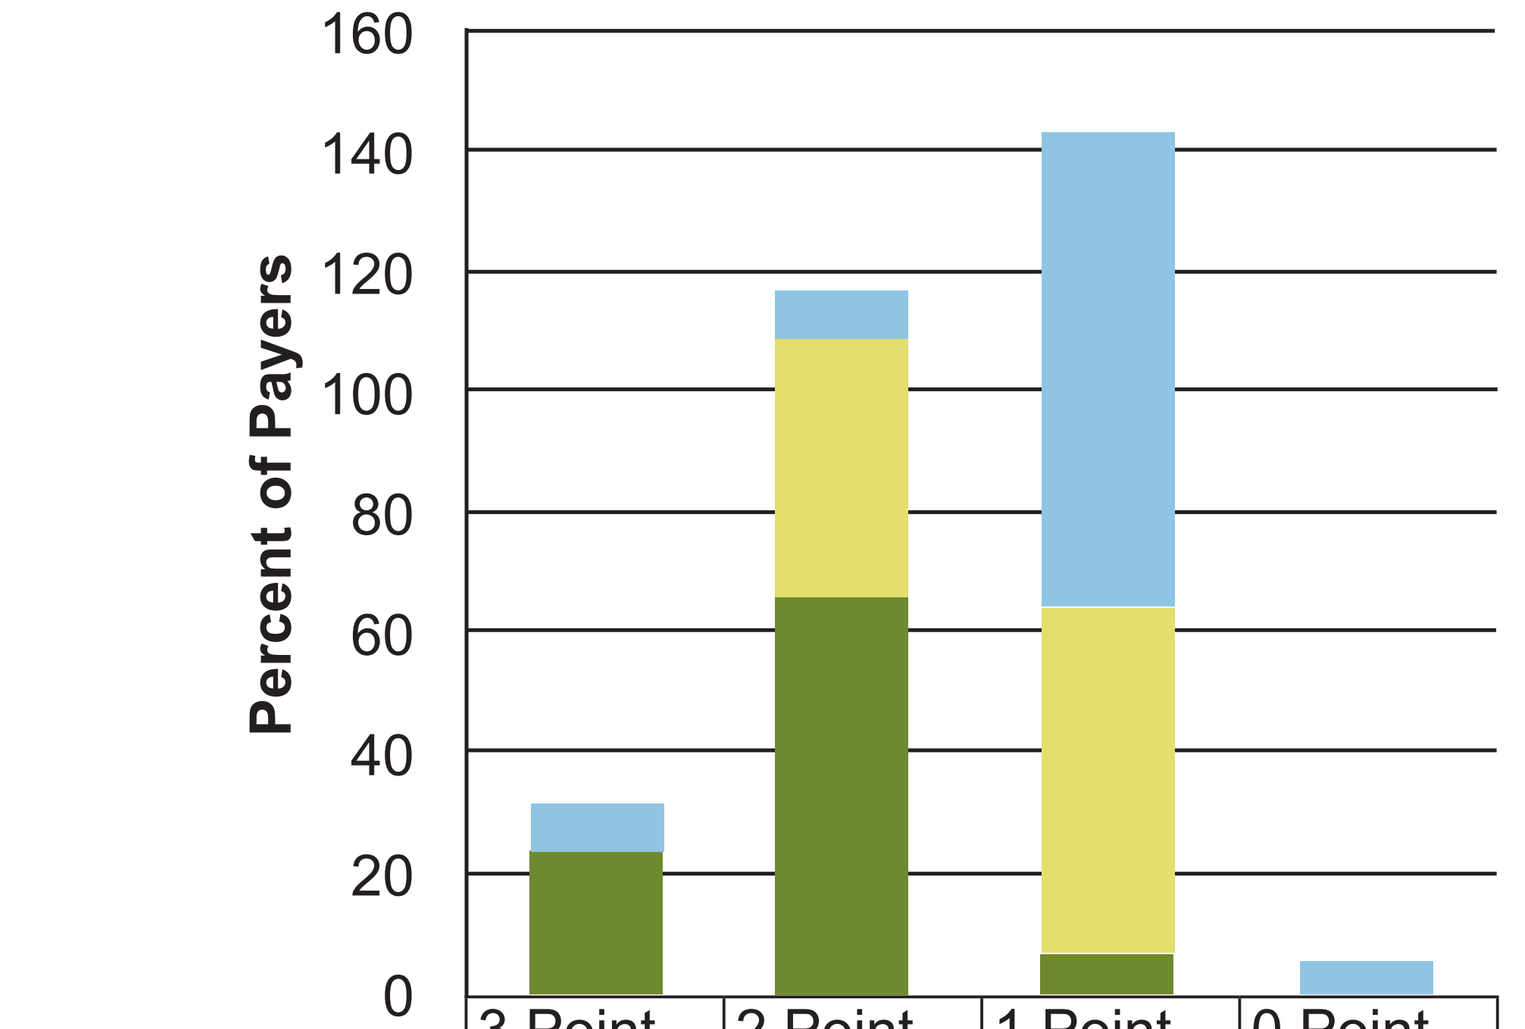
Percent of Payers

(n=12)	3 Point Scale	2 Point Scale	1 Point Scale	0 Point Scale
Influence	8% (1)	8% (1)	75% (9)	8% (1)
Completeness	0% (0)	42% (5)	58% (7)	0% (0)
Overall Value	25% (3)	67% (8)	8% (1)	0% (0)

Table 4. Mean Point Scale of Payers on Section 3

Influence	1.17
Completeness	1.42
Overall Value	2.17

Table 5. Time Invested by Manufacturers on Section 3 (mean)

Preparing (n=10)	66 hrs (range 5-168)
Updating (n=10)	26 hrs (range 3-60)
Combined	46 hrs

Figure 14. Overall Payers' Opinions of Section 4

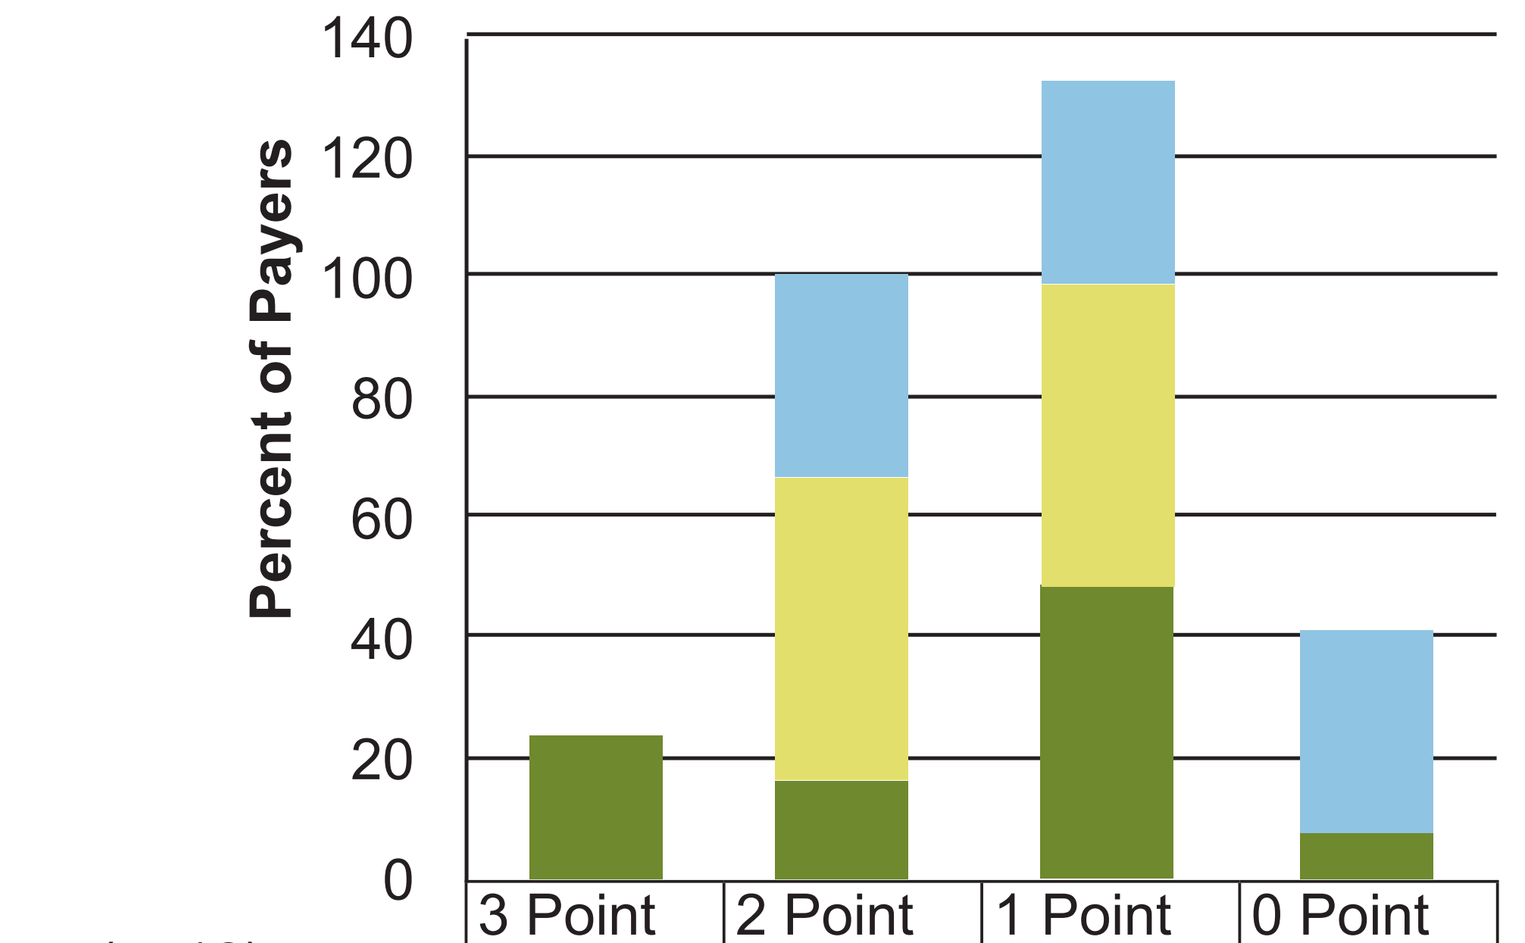
Percent of Payers

(n=12)	3 Point Scale	2 Point Scale	1 Point Scale	0 Point Scale
Influence	0% (0)	33% (4)	33% (4)	33% (4)
Completeness	0% (0)	50% (6)	50% (6)	0% (0)
Overall Value	25% (3)	17% (2)	50% (6)	8% (1)

Table 6. Mean Point Scale of Payers on Section 4

Influence	1.00
Completeness	1.50
Overall Value	1.58

Table 7. Time Invested by Manufacturers on Section 4 (mean)

Preparing (n=4)	39 hrs (range 6-120)
Updating (n=4)	9 hrs (range 2-20)
Combined	24 hrs

- 83% (10/12) payers valued the Supporting Clinical Evidence section over the Economic Value and Modeling Report section.

Table 8. Reasons Payers Value Section 3 Over Section 4

- Clinical evidence is universal, while economic value is sometimes plan specific, based on area and population.
- Clinical evidence does not change.
- Economic value and model reports can be manipulated by different considerations and assumptions given.
- Each health system has a unique financial structure that makes it difficult to use models.
- Payers can perform their own economic analysis to fit their needs.
- Data and evidence is more concrete than marketing projections.
- Formulary decisions are based solely on efficacy and safety data for a drug.

DISCUSSION

- In the last five years, there has been an increasing trend of dossier requests.
- The majority of payers found the clinical evidence more valuable than the economic evidence, which may be why manufacturers invested more time on Section 3.
 - There were conflicting opinions on unpublished studies for labeled and off-labeled indications.
 - About half as much time was needed to prepare and update Section 4 compared to Section 3.
- Payers and manufacturers agreed that cost-effectiveness models were the most valued and more consistently included but had differing opinions on the modeling technique used to construct cost-effectiveness models.
 - Models were not as user-friendly or clear as payers would have liked.
 - Payers felt that models were only somewhat transparent, whereas manufacturers believed their models were transparent to most, if not all, health systems.
 - AMCP recommends the implementation of models into a spreadsheet software to enhance transparency and ease of use, but the software was used by only two-thirds of manufacturers.

LIMITATIONS

- It is difficult to generalize these results to payers and manufacturers outside the sample population due to the small sample size.
- Several payers were not recruited for this study because they were neither in the PBM Institute Directory nor had a managed care pharmacy residency program.
- The manufacturer survey was only sent to one MI representative for each company but contained multiple economic questions that may have extended beyond the responsibilities of the MI department (i.e. Outcomes Research).
- The assumption was made that information consistently included in the dossier and greater time invested by manufacturers on the dossier was related to value.
- Two payers did not review economic models, but their responses were included in the payers' overall opinion analysis of Section 4.
- Due to the anonymous nature of these surveys, it was possible for more than one respondent to be from the same company.
- This study was the first of its kind and took an innovative approach to assess dossier value, though a validated scale was not used.

CONCLUSION

- The results from both the payer and pharmaceutical manufacturer surveys provided insight to manufacturers as to which type of evidence-based information is more valuable and helped identify whether current dossier preparation methods meet the needs of payers.
- Improvements payers would like to see in dossier submissions:
 - Head-to-head comparison studies
 - Transparency in models
 - Balanced interpretation of clinical evidence
 - Less variability in quality
 - Unpublished studies for off-labeled indications
- Medical Information, Medical Affairs, and Outcomes Research departments can use this data to re-evaluate the type of information for dossier inclusion and reassess the allocation of time and resources for dossier development.
- Manufacturers may benefit by working closely with individual payers to develop customized dossiers that are in line with the specific areas of value to each payer.

REFERENCES

1. Sullivan SD, Lyles A, Luce B, et al. AMCP Guidance for submission of clinical and economic evaluation data to support formulary listing in US health plans and pharmacy benefits management *organizations. J Managed Care Pharm*. 2001;7(4):272–82
2. Academy of Managed Care Pharmacy. The AMCP format for formulary submissions version 3.0. October 2009. Available at: http://www.amcp.org/format/pub.pdf. Accessed December 13, 2010.
3. Neumann PJ. Evidence-based and value-based formulary guidelines. *Health Affairs.* 2004;23(1): 124-34.
4. Colmernero F, Sullivan SD, Palmer JA, et al. Quality of clinical and economic evidence in dossier formulary submissions. *Am J Manag Care*. April 2006;16(7):401-7.
5. Watkins JB, Minshall ME, Sullivan SD. Application of economic analyses in US managed care formulary decisions: a private payer's experience. *J Manag Care Pharm*. 2006;12(9):726-35.
6. Olson BM, Armstrong EP, Grizzle AJ, et al. Industry's perception of presenting pharmacoeconomic models to managed care organizations. *J Manag Care Pharm*. 2003;9(2):159-67.
7. Spooner JJ, Gandhi PK, Connelly SB. AMCP format dossier requests: manufacturer response and formulary implications for one large health plan. *J Manag Care Pharm*. 2007;13(1):37-43.
8. Nichol MB, Knight TK, Epstein J, et al. Opinions regarding the Academy of Managed Care Pharmacy dossier submission guidelines: results of a small survey of managed care organizations and pharmaceutical manufacturers. *J Manag Care Pharm*. May 2007;13(4):360-71.

Philadelphia College of Pharmacy University of the Sciences

Centocor Ortho Biotech Services LLC